Fingolimod: long-term (up to 4 years) efficacy in patients with relapsing-remitting multiple sclerosis in FREEDOMS and FREEDOMS II extension studies

DX 25

Douglas Jeffery,[1] Anthony T. Reder,[2] Xiangyi Meng,[3] Simrat Randhawa[3]

[1]Cornerstone Healthcare, High Point, NC; [2]University of Chicago Medical Center, Chicago, IL; [3]Novartis Pharmaceutical Corporation, East Hanover, NJ

CONCLUSIONS

- **Results from FREEDOMS and FREEDOMS II extension studies demonstrated sustained (up to 4 years) clinical and MRI benefits in patients with relapsing MS and support the long-term efficacy of fingolimod.**
- **After switching from placebo to fingolimod, patients experienced improvements in ARR similar to those observed in patients receiving fingolimod continuously from study initiation.**
 - **Clinical benefits were seen with continuous fingolimod treatment relative to core study results as well as in patients who switched to fingolimod from placebo.**
- **In both studies, continuous treatment with fingolimod was associated with reduced brain atrophy vs switching to fingolimod at month 24, implying a smaller overall volume loss with earlier initiation of fingolimod.**

INTRODUCTION

- Fingolimod is a once-daily, oral sphingosine 1-phosphate receptor modulator approved in the United States and European Union for the treatment of relapsing multiple sclerosis (MS).[1,2a]
- In the 24-month FREEDOMS (NCT00289978) and FREEDOMS II (NCT00355134) core studies, once-daily fingolimod (0.5 or 1.25 mg) showed beneficial effects on clinical and magnetic resonance imaging (MRI) endpoints vs placebo in patients with relapsing-remitting MS.[3,4]

OBJECTIVE

- To report long-term efficacy outcomes (up to 4 years) in patients treated with fingolimod in the FREEDOMS and FREEDOMS II extension studies

METHODS

Study Design and Assessments

- The FREEDOMS and FREEDOMS II core studies were phase 3, double-blind, placebo-controlled trials including patients 18–55 years of age with relapsing-remitting MS, ≥1 relapse in the previous year (or ≥2 in the previous 2 years), and an Expanded Disability Status Scale score of 0–5.5.[3,4]
- The FREEDOMS and FREEDOMS II extension trials included patients who completed the individual core studies.
- In the extension trials, patients who were treated with fingolimod 0.5 or 1.25 mg during the core study continued the same dose (continuous group), and those who were treated with placebo were rerandomized 1:1 to fingolimod 0.5 or 1.25 mg (switch group).
 - After a study amendment was made to discontinue fingolimod 1.25 mg, all patients received open-label fingolimod 0.5 mg until the end of study (EOS).
- The extension trials continued until the umbrella safety extension study (NCT01201356) opened enrollment to all patients who completed selected ongoing or planned trials in the fingolimod clinical development program.
- Key efficacy endpoints included annualized relapse rate (ARR) and changes in MRI measures of disease activity.

Statistical Analyses

- Between-group comparisons were based on the core intent-to-treat (ITT) population to evaluate outcomes in patients continuously treated (0.5 and 1.25 mg) vs patients who were switched (placebo-merged fingolimod 0.5/1.25 mg).
- Within-group comparisons were based on the extension ITT population to compare treatment effects of fingolimod 0.5 mg in the extension phase vs the core phase.
 - For FREEDOMS, comparisons were made for months 0–24 vs months 24–48.
 - For FREEDOMS II, the number of patients with available data at month 48 was low (n=54); therefore, comparisons were limited to months 24–36 vs months 0–12 and 12–24.
- Tests of hypotheses were 2-sided at a significance level of 0.05, with no adjustment for multiplicity.

[a]*The approved indication may vary from country to country. In the United States, fingolimod is approved for the treatment of patients with relapsing forms of MS. In the European Union, fingolimod is approved for treatment of patients with highly active relapsing-remitting MS.*

RESULTS

Patient Disposition

- Of 1272 patients randomized in the FREEDOMS core study, 920 (72.3%) entered the extension.
 - Of these patients, 773 (84.0%) completed the study, 581 (63.2%) were continuously treated with fingolimod for 3 years, and 277 (30.1%) were continuously treated for 4 years.
- Of the 1083 patients randomized in the FREEDOMS II core study, 632 (58.4%) entered the extension.
 - Of these patients, 529 (83.7) completed the study, 212 (33.5%) were continuously treated with fingolimod for ≥3 years, and 54 (8.5%) had data available at 4 years.

ARR

- Between-group comparisons in each study showed continuous fingolimod significantly reduced ARR from month 0 to EOS vs switching from placebo (all *P*<0.001; **Figure 1**).

Figure 1. Between-group comparisons of ARR (month 0 to EOS) in (A) FREEDOMS and (B) FREEDOMS II*

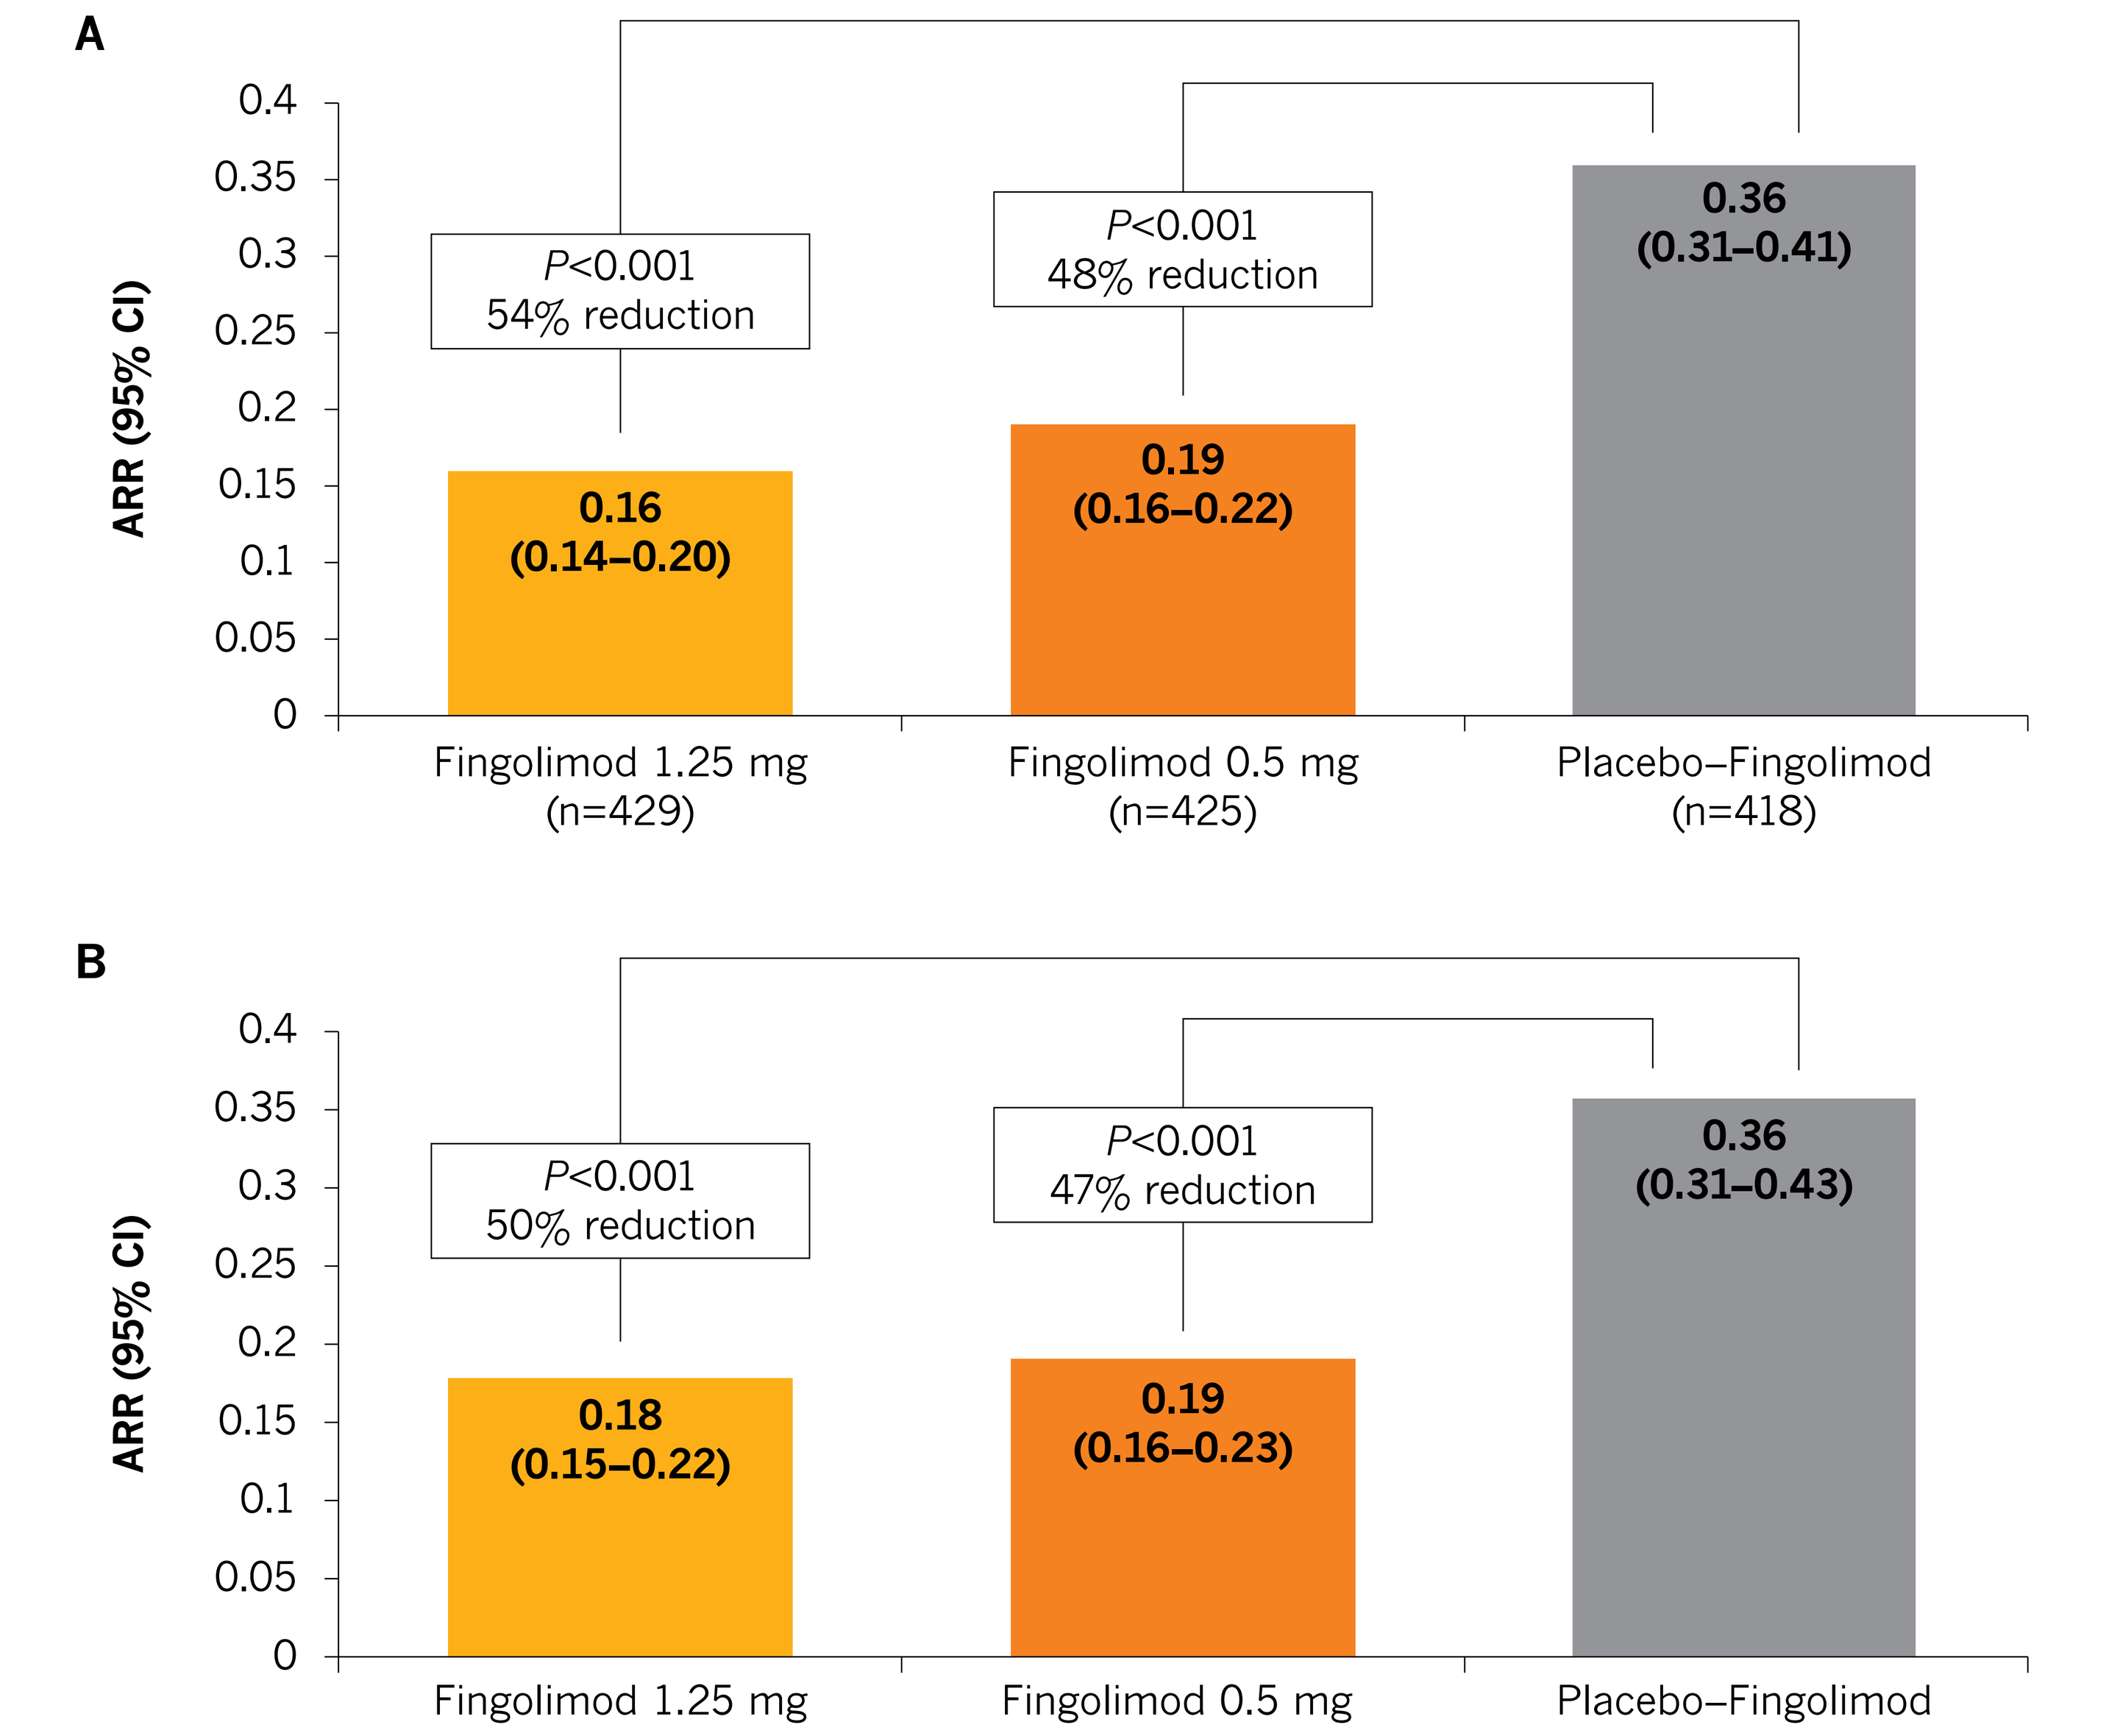
A
ARR (95% CI)
0.4
0.35
0.3
0.25
0.2
0.15
0.1
0.05
0
P<0.001
54% reduction
P<0.001
48% reduction
0.16
(0.14–0.20)
0.19
(0.16–0.22)
0.36
(0.31–0.41)
Fingolimod 1.25 mg
(n=429)
Fingolimod 0.5 mg
(n=425)
Placebo–Fingolimod
(n=418)
B
P<0.001
50% reduction
P<0.001
47% reduction
0.18
(0.15–0.22)
0.19
(0.16–0.23)
0.36
(0.31–0.43)
Fingolimod 1.25 mg
Fingolimod 0.5 mg
Placebo–Fingolimod

ARR=annualized relapse rate; EOS=end of study; ITT=intent to treat.
*Analyses based on core ITT populations.

- Within-group comparisons showed ARR remained low in continuous fingolimod 0.5 mg groups, and patients who switched from placebo to fingolimod 0.5 mg had an approximate 50% reduction in ARR during the extension (*P*<0.05 vs core phase in both studies; **Figure 2**).

Figure 2. Within-group comparison of ARR in core vs extension phases in (A) FREEDOMS and (B) FREEDOMS II*

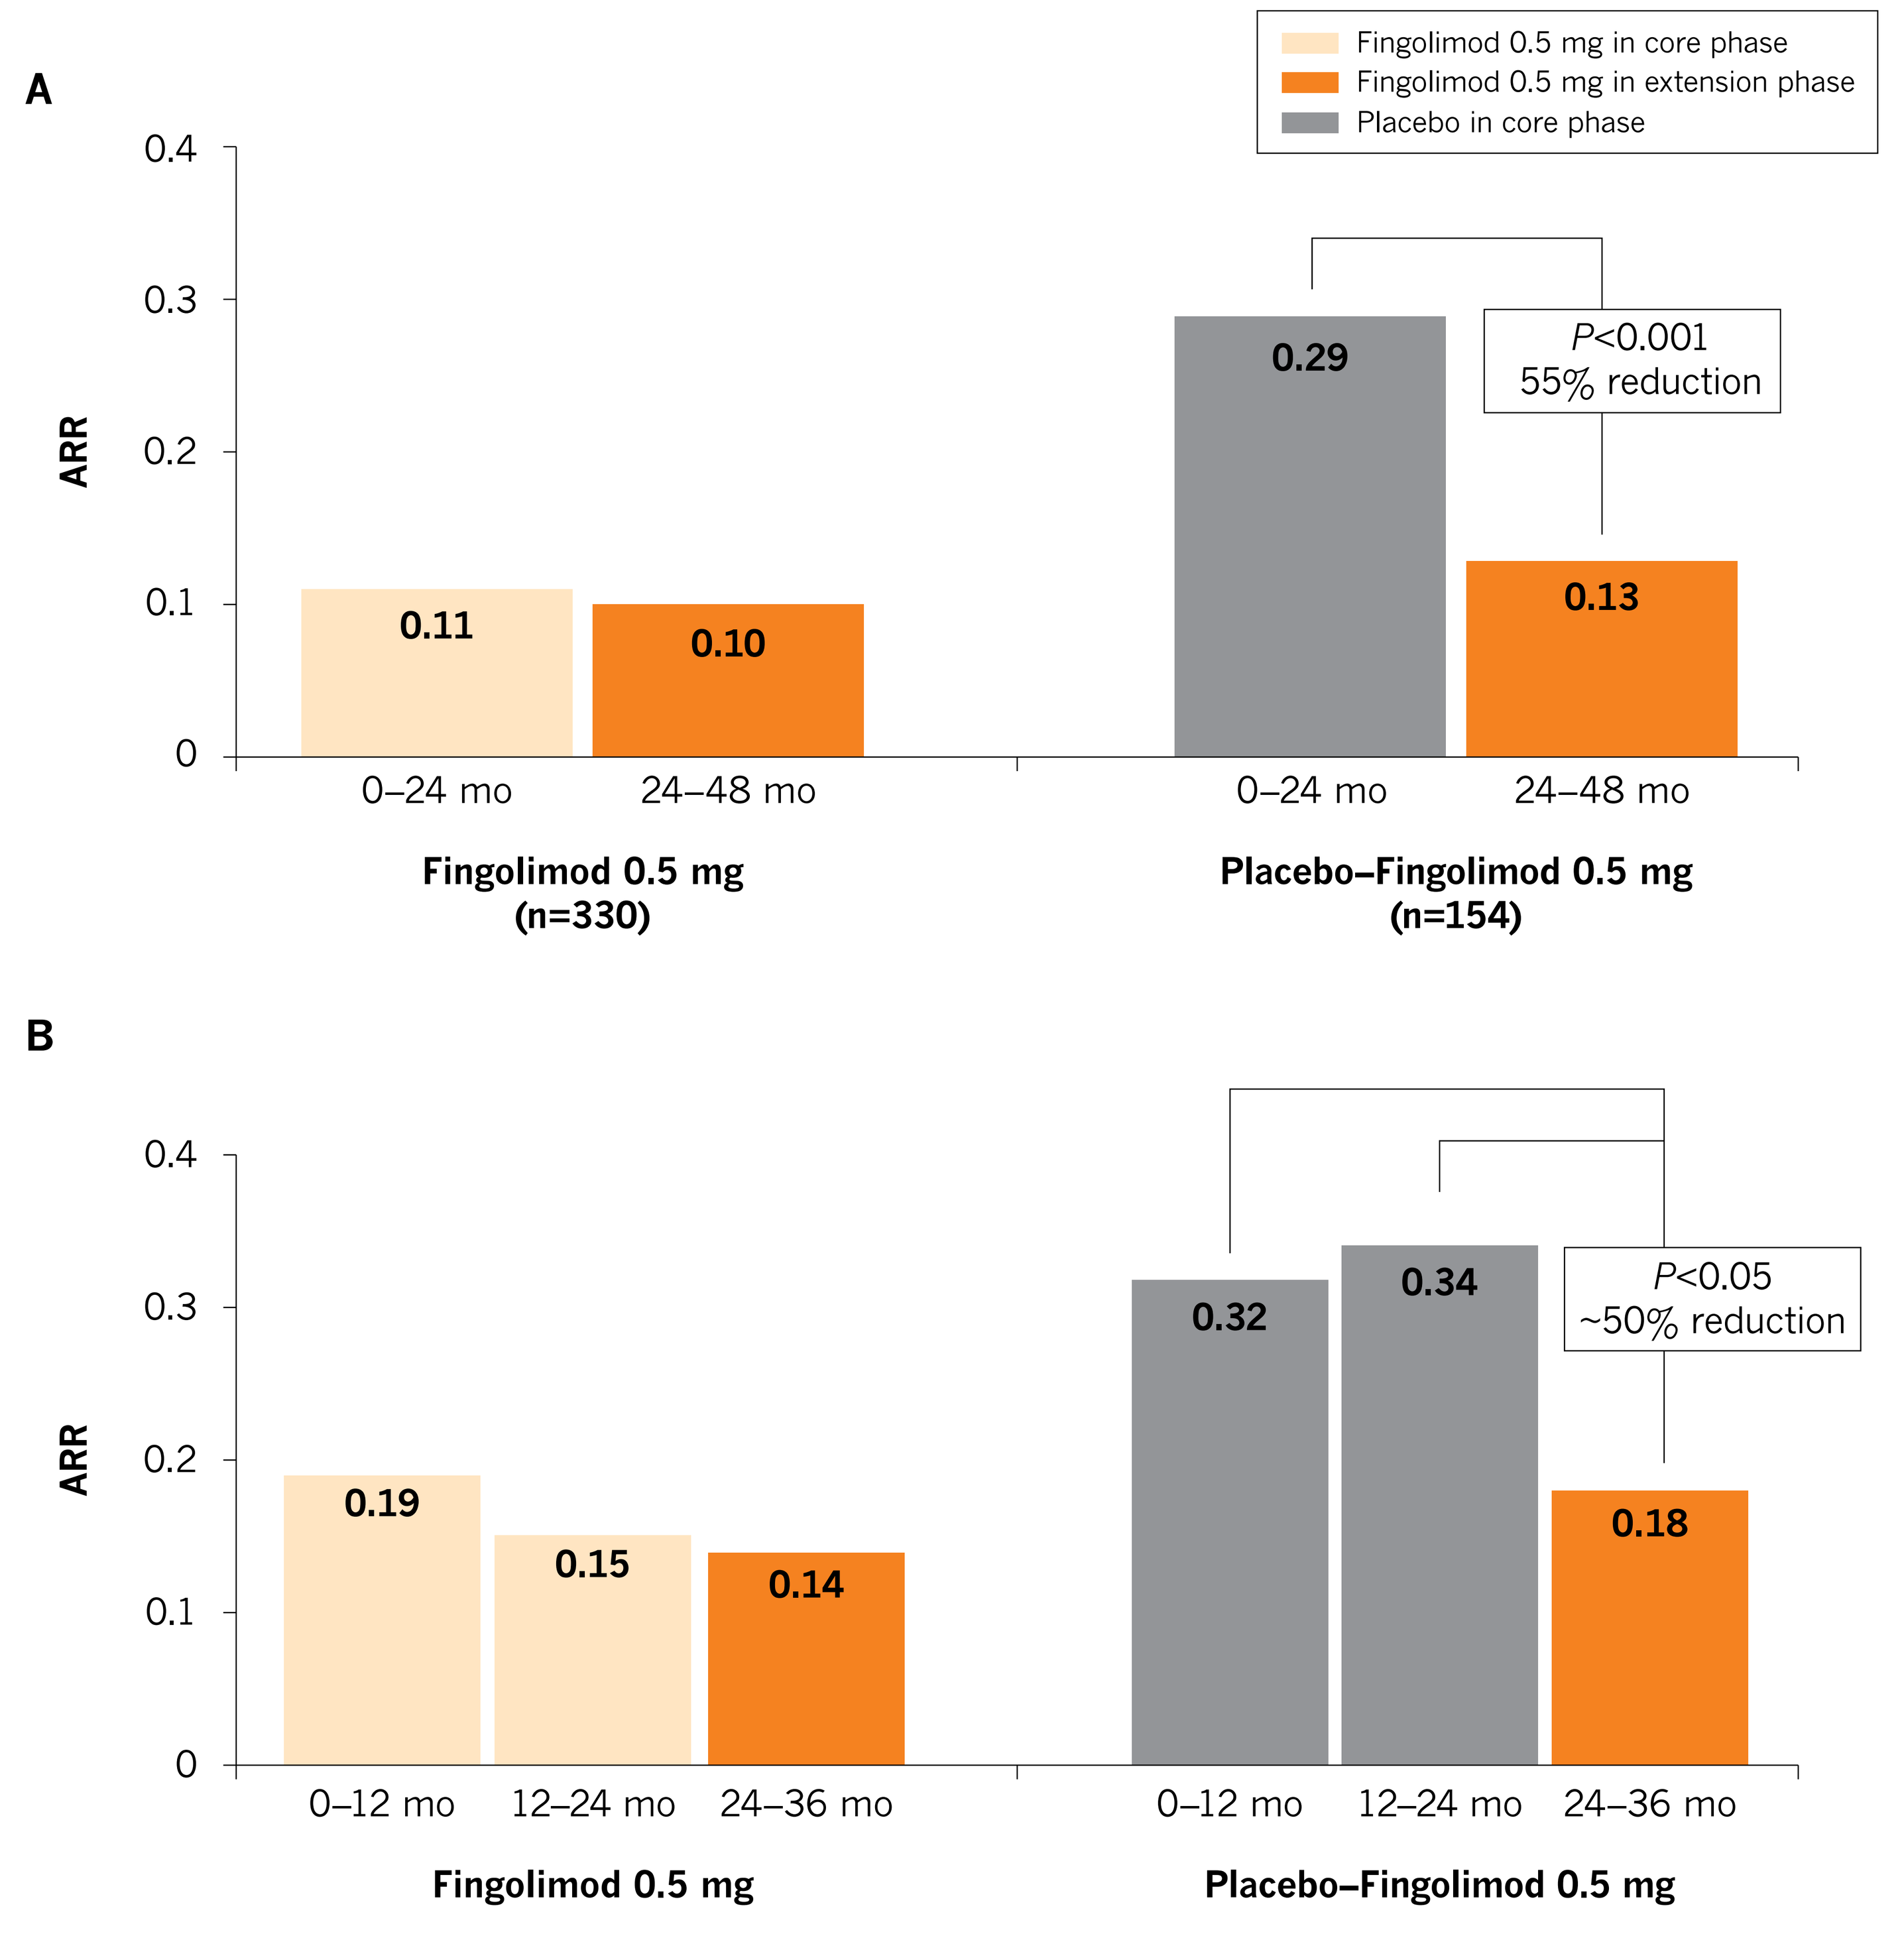
Fingolimod 0.5 mg in core phase
Fingolimod 0.5 mg in extension phase
Placebo in core phase
A
ARR
0.4
0.3
0.2
0.1
0
0.11
0.10
0.29
P<0.001
55% reduction
0.13
0–24 mo
24–48 mo
0–24 mo
24–48 mo
Fingolimod 0.5 mg
(n=330)
Placebo–Fingolimod 0.5 mg
(n=154)
B
0.19
0.15
0.14
0.32
0.34
P<0.05
~50% reduction
0.18
0–12 mo
12–24 mo
24–36 mo
0–12 mo
12–24 mo
24–36 mo
Fingolimod 0.5 mg
Placebo–Fingolimod 0.5 mg

ARR=annualized relapse rate; ITT=intent to treat.
*Analyses based on extension ITT populations.

MRI Measures of Disease Activity

- MRI measures showed significantly reduced brain atrophy in the continuous fingolimod groups vs the switch group at EOS in both studies (all *P*≤0.006; **Figure 3**).

Figure 3. Between-group comparison of mean percentage change in brain volume from month 0 to EOS in (A) FREEDOMS and (B) FREEDOMS II*

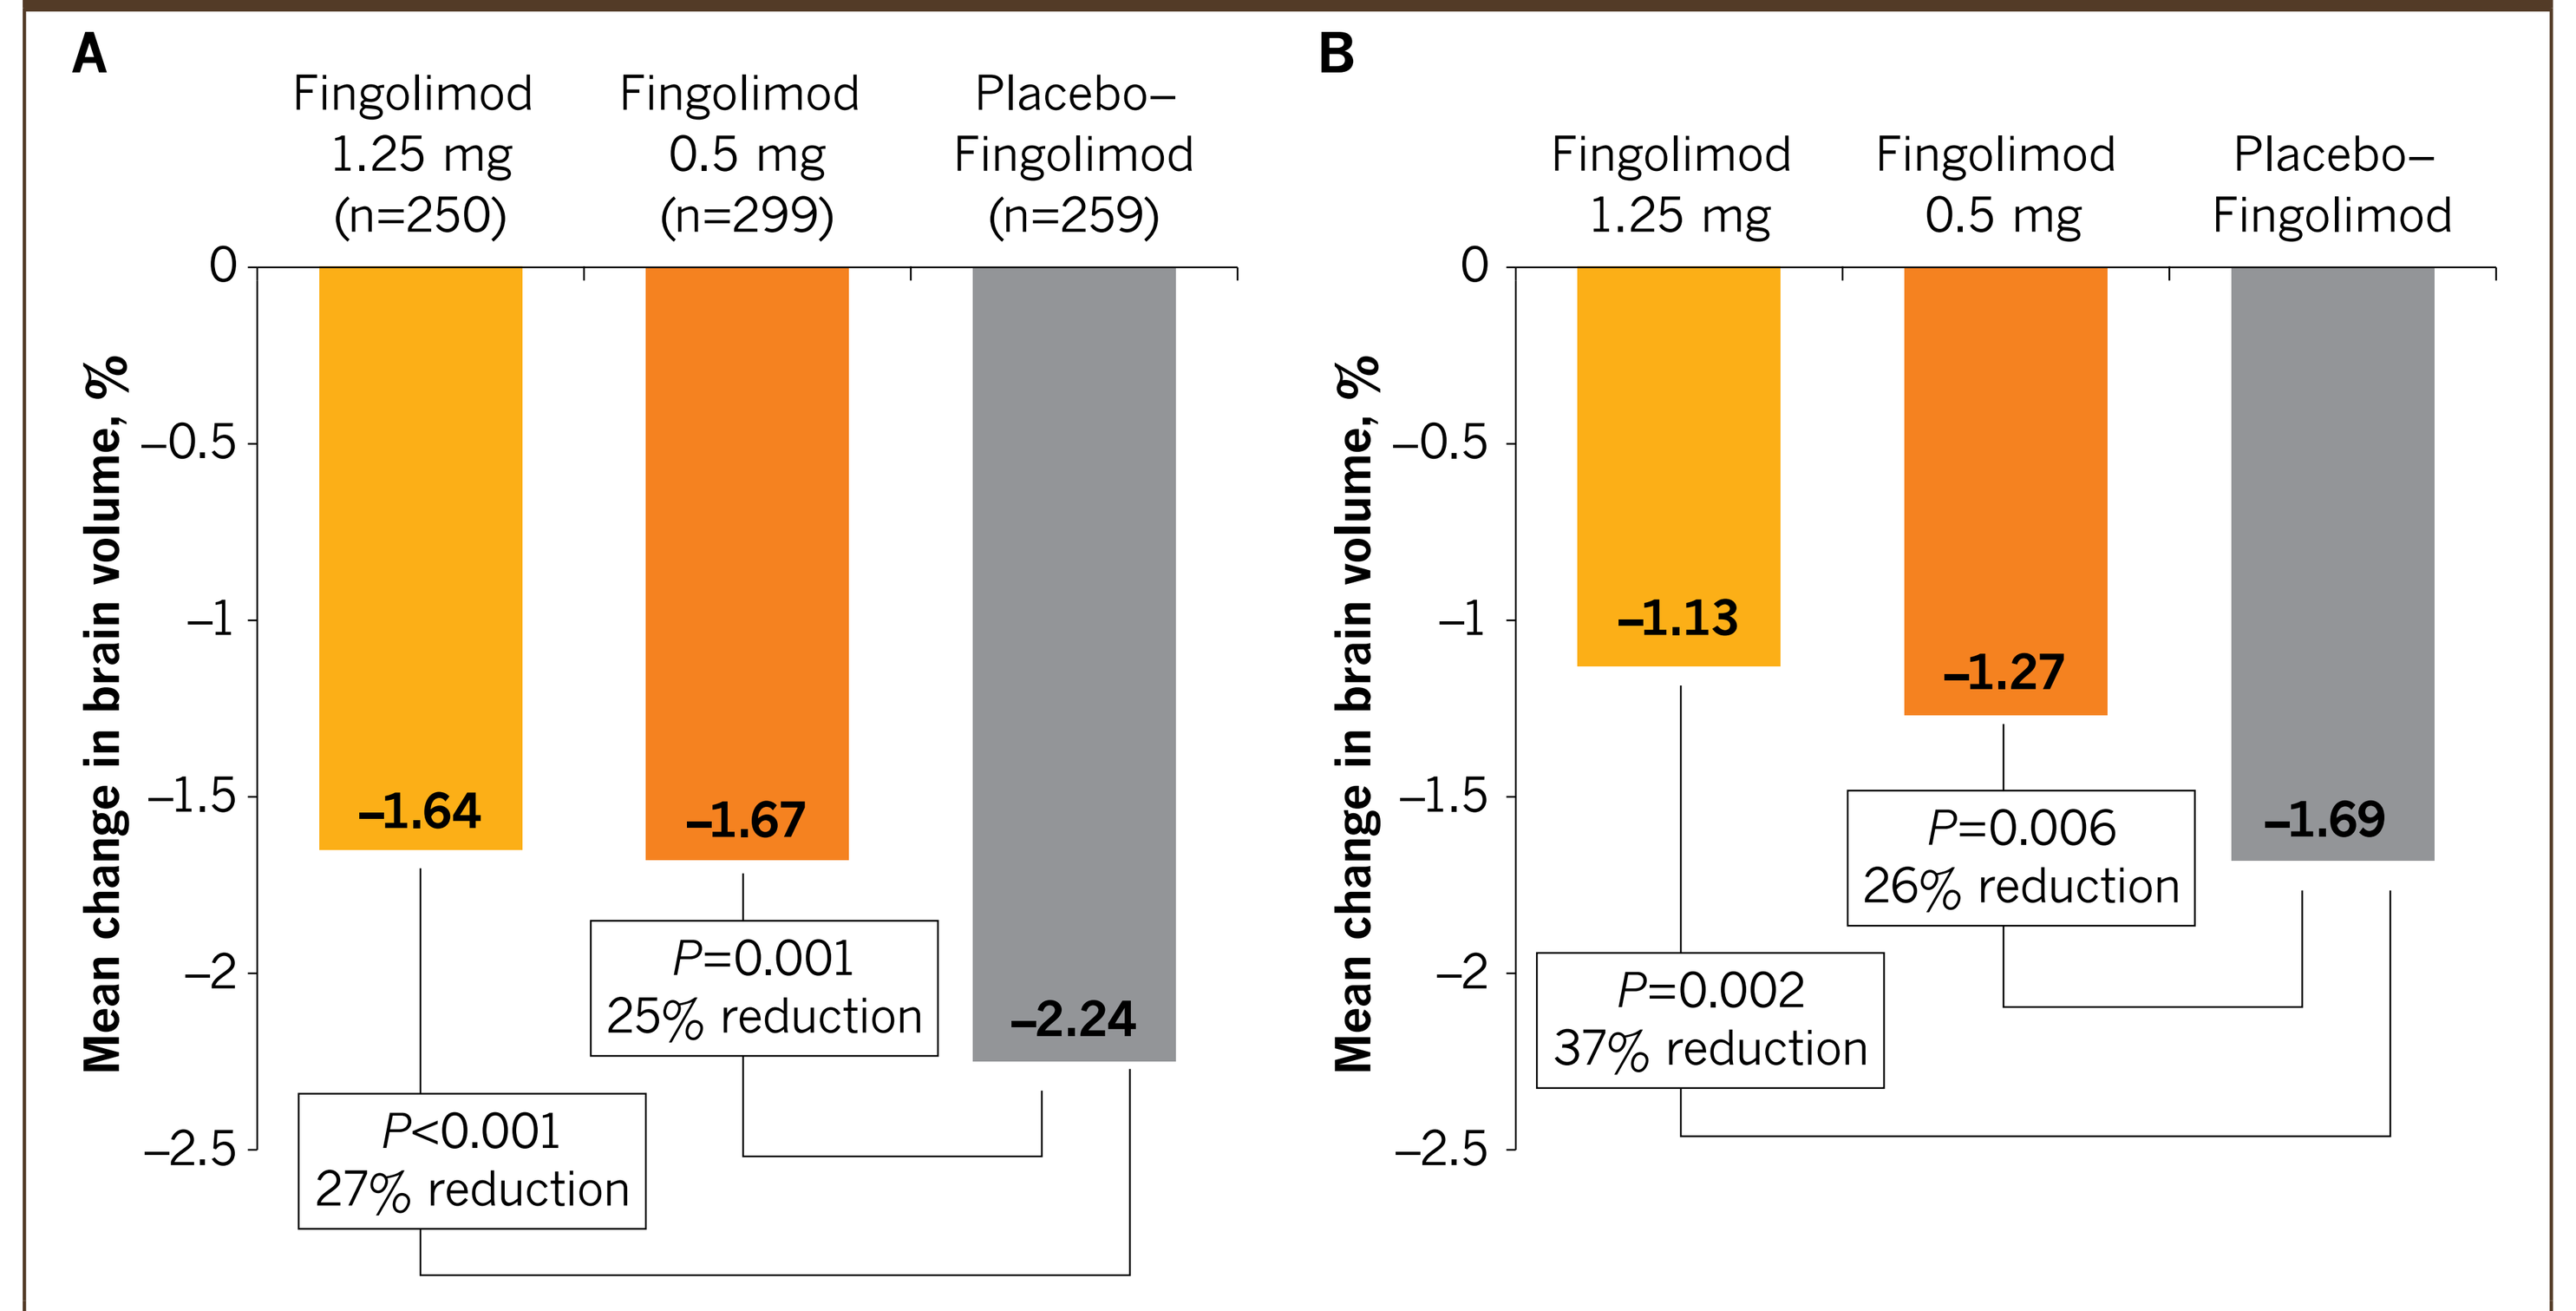
A
Fingolimod
1.25 mg
(n=250)
Fingolimod
0.5 mg
(n=299)
Placebo–
Fingolimod
(n=259)
Mean change in brain volume, %
0
−0.5
−1
−1.5
−2
−2.5
–1.64
–1.67
–2.24
P=0.001
25% reduction
P<0.001
27% reduction
B
Fingolimod
1.25 mg
Fingolimod
0.5 mg
Placebo–
Fingolimod
–1.13
–1.27
–1.69
P=0.006
26% reduction
P=0.002
37% reduction

EOS=end of study; ITT=intent to treat.
*Analyses based on core ITT populations.

- In FREEDOMS, patients who switched to fingolimod had significantly reduced brain atrophy in the extension phase vs the core phase (mean % change in brain volume: –0.9% vs –1.42% with placebo in the core phase; *P*=0.008).
 - In FREEDOMS II, decreases in brain volume from month 24 to EOS were not statistically different across all treatment groups.

References

1. Gilenya® (fingolimod). Full Prescribing Information, Novartis Pharmaceuticals Corporation, East Hanover, NJ, 2012.
2. Gilenya® (fingolimod). Summary of Product Characteristics. Novartis Pharmaceuticals UK Ltd, Camberley, Surrey, UK. Available at: http://www.medicines.org.uk/emc/medicine/24443. Accessed May 13, 2014.
3. Kappos L, et al. *N Engl J Med.* 2010;362:387-401.
4. Calabresi PA, et al. Safety and efficacy of fingolimod in patients with relapsing-remitting multiple sclerosis (FREEDOMS II): a double-blind, randomised, placebo-controlled, phase 3 trial. *Lancet Neurol.* 2014:[Epub ahead of print].

Disclosures

D. Jeffery has served as a consultant for Bayer, Biogen Idec, Teva, EMD Serono, Pfizer, GlaxoSmithKline, Novartis, Acorda, Genzyme, and Questcor and received grant/research support from Bayer, Biogen Idec, Teva, EMD Serono, Pfizer, and Novartis.

A. T. Reder has served as a consultant for, has received unrestricted research grants from, and has performed clinical trials funded by Novartis.

X. Meng and S. Randhawa are employees of Novartis.

Acknowledgments

This study was supported by Novartis Pharmaceuticals Corporation. The authors thank Meg Church, MS, and Erica Wehner, RPh, CMPP, for medical writing support of this poster. Editorial support was provided by Complete Healthcare Communications, Inc. (Chadds Ford, PA). Medical writing and editorial support were funded by Novartis.

Scan to download a reprint of this poster

Downloading data may incur costs which can vary depending on your service provider and may be high if you are using your smartphones abroad. Please check your phone tariff or contact your service provider for more details.

This study was supported by Novartis Pharmaceuticals Corporation | Poster presented at the 2014 Annual Meeting of the Consortium of Multiple Sclerosis Centers (CMSC) and the Sixth Cooperative Meeting with Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS); May 28–31, 2014; Dallas, TX.