DX58

Teriflunomide in Routine Clinical Practice: Design of the Teri-PRO Study

Timothy Vollmer MD,[1] Aline Stalla-Bourdillon MD,[2] Miqun Robinson, MD[3]

[1]University of Colorado Health Sciences Center, Aurora, USA; [2]Genzyme, a Sanofi company, Chilly-Mazarin, France; [3]Genzyme, a Sanofi company, Bridgewater, NJ, USA

BACKGROUND

- Teriflunomide is a once-daily oral immunomodulator approved in the USA, Australia, and Argentina for the treatment of relapsing forms of multiple sclerosis (RMS).
- Teriflunomide selectively and reversibly inhibits dihydro-orotate dehydrogenase, a key mitochondrial enzyme in de novo pyrimidine synthesis required for rapidly dividing lymphocytes.[1, 2]
 - Through this cytostatic effect, teriflunomide limits expansion of stimulated T and B cells thought to be responsible for the damaging inflammatory process associated with MS.[1, 2]
 - Basic homeostatic cell functions of resting and slowly dividing cells appear to be preserved, so lymphocytes remain available for ongoing immune surveillance.[3]
- The efficacy and safety of teriflunomide has been investigated in an extensive clinical development program and it has shown efficacy in the two pivotal phase 3 trials, TEMSO (**TE**riflunomide **M**ultiple **S**clerosis **O**ral: NCT00134563) and TOWER (**T**eriflunomide **O**ral in people **W**ith relapsing multipl**E** scle**R**osis: NCT00751881).[4, 5]
- In both studies, teriflunomide 14 mg significantly reduced annualized relapse rate compared with placebo (31.5%, p<0.001 in TEMSO and 36.3%, p<0.001 in TOWER). A significant decrease was also observed with teriflunomide 14 mg in sustained disability progression (confirmed for 12 weeks) compared with placebo (29.8%, p=0.028 in TEMSO and 31.5%, p=0.044 in TOWER). Once-daily treatment with teriflunomide 7 mg also significantly reduced annualized relapse rate in both studies (31.2%, p<0.001 in TEMSO and 22.3%, p=0.002 in TOWER), but not disability progression.[4, 5]
- Patient-reported outcomes (PROs) provide clinical information to support the evaluation of MS therapies. Patients reported only small changes from baseline in health-related quality of life (HR-QoL) outcomes (EuroQol-5D and Short-Form 36) with no significant differences among the study groups in TEMSO and TOWER.[4, 6, 7] In the comparative trial with interferon beta 1-a (IFN β-1a; TENERE: NCT00883337), patients expressed greater global satisfaction with treatment (related to effectiveness, side-effects, and convenience) in the teriflunomide 7 mg (p=0.024) and 14 mg (p=0.016) groups than in the IFN β-1a group, as assessed by the Treatment Satisfaction Questionnaire for Medication (TSQM, secondary outcome).[8]
- The phase 4 Teri-PRO (**Teri**flunomide **P**atient-**R**eported **O**utcomes) study will further investigate patient perceptions of teriflunomide on treatment satisfaction, QoL, and effectiveness using PROs, and will provide valuable information on patient adherence and persistence with teriflunomide therapy in real-life settings.

STUDY OBJECTIVES

Primary Objective

- To describe the efficacy, tolerability, and convenience of teriflunomide treatment through the evaluation of PROs.

Key Secondary Objectives

- To describe disease progression using PROs.
- To describe clinical outcomes (ie, treated relapses, time to first relapse).
- To describe change in cognition.
- To describe safety and tolerability (based on adverse event [AE] reporting, vital signs evaluation, and laboratory testing).
- To describe adherence (ie, compliance) and persistence (ie, duration of exposure) to teriflunomide treatment.
- To describe changes in QoL, leisure, and activity.
- To compare Patient-Determined Disease Steps (PDSS) and the Expanded Disability Status Scale (EDSS) in assessing MS disease progression.

METHODS

Study Design

- Teri-PRO is a prospective, single-arm, multicenter, real-life study to describe the efficacy, tolerability, and convenience of teriflunomide treatment using PROs in patients with RMS who are either naïve to disease-modifying treatment (DMT) or switching from another DMT. Teriflunomide will be provided.
- A maximum of 1000 patients across 200 sites will receive teriflunomide once-daily for 48 weeks, with doses given according to local labeling.
- The study design is shown in **Figure 1**.

Figure 1. Teri-PRO Study Design

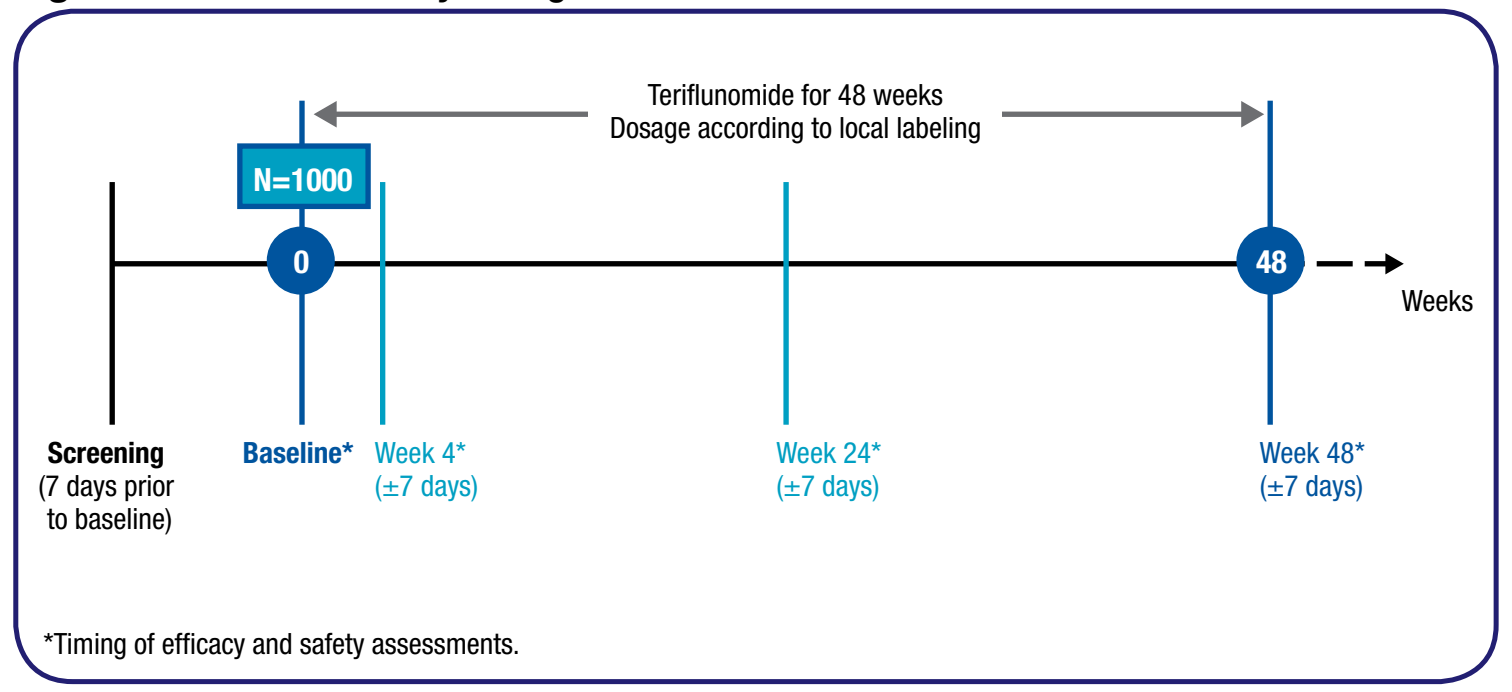
Teriflunomide for 48 weeks
Dosage according to local labeling
N=1000
0
48
Weeks
Screening
(7 days prior to baseline)
Baseline*
Week 4*
(±7 days)
Week 24*
(±7 days)
Week 48*
(±7 days)
*Timing of efficacy and safety assessments.

Patient Entry Criteria

- The key inclusion and exclusion criteria for the Teri-PRO study are shown in **Table 1**.

Table 1. Inclusion and Key Exclusion Criteria for the Teri-PRO Study

Inclusion criteria	• Patients with relapsing forms of MS, considered to have indication for teriflunomide treatment • Signed written informed consent
Exclusion criteria	• Under 18 years of age • Current or history of treatment with teriflunomide • Previous exposure to leflunomide within 6 months prior to baseline • Pre-existing acute or liver disease, or those with serum alanine aminotransferase ≥2 times the upper limit of normal • Known history of severe immunodeficiency, bone marrow disease, acute or severe active infections • Pregnancy or breastfeeding • Unwilling to use reliable contraception

Study Endpoints

Primary Endpoint

- Assessment at 48 weeks (end of treatment [EOT]) of global satisfaction with teriflunomide treatment, measured by score on TSQM, version 1.4 (**Table 2** and **Table 3**).

Table 2. Patient-Reported Outcome Measures Used in Teri-PRO

Treatment Satisfaction Questionnaire for Medication (TSQM) version 1.4[9]	• To assess overall level of satisfaction or dissatisfaction of patients with their medication • Consists of 14 questions focusing on effectiveness, side-effects, and convenience of the medication over the past 2–3 weeks, or since patient's last use
Patient-Determined Disease Steps (PDDS)[10]	• To assess disability in patients with MS; PDDS focuses mainly on how patients walk • Consists of a scale scored as: 0, normal; 1, mild disability; 2, moderate disability; 3, gait disability; 4, early cane; 5, late cane; 6, bilateral support; 7, wheelchair/scooter; and 8, bedridden
Multiple Sclerosis Performance Scale (MSPS)[11]	• To assess disability in patients with MS • Patients indicate the category that best describes their condition during the past month on 11 subscales: Mobility, Hand Function, Vision, Fatigue, Cognitive, Bladder/Bowel, Sensory, Spasticity, Pain, Depression, and Tremor and Coordination • All of the subscales are scored as 0, normal; 1, minimal; 2, mild; 3, moderate; 4, severe; or 5, total disability (except Mobility, which is scored from 0 to 6)
Multiple Sclerosis International Quality of Life (MusiQoL)[12]	• To assess quality of life in patients with MS • List of 31 questions specific for use in patients with the following forms of MS: relapsing–remitting, secondary progressive, progressive relapsing, and clinically isolated syndrome
Stern Leisure Activity Scale[13]	• To assess cognition and quality of life • List of 12 questions assessing the frequency of leisure activities in the past month

Table 3. Efficacy and Safety Assessments

	Screening	Treatment Period			
VISIT	V1 (Screening)	V2 (Baseline)	V3	V4	V5 or premature EOT
DAY	D −7 to D1	D1	W4± 7D	W24± 7D	W48± 7D
General					
Informed consent	✓				
Accelerated elimination procedure					✓
Efficacy					
TSQM		✓	✓		✓
MSPS		✓		✓	✓
PDDS		✓			✓
Relapses		✓	✓	✓	✓
EDSS		✓			✓
SDMT		✓			✓
MusiQoL		✓			✓
Stern Leisure Activities scale		✓			✓
Treatment					
Dispense IMP		✓	✓	✓	
Adherence to study treatment			✓	✓	✓
Safety					
AEs/SAEs recording (if any)		✓	✓	✓	✓
Vital signs (including blood pressure)		✓	✓	✓	✓
Pregnancy test	✓				
CBC	✓				✓
Liver function (ALT, AST, bilirubin)	✓		✓	✓	✓

AE, adverse event; ALT, alanine aminotransferase; AST, aspartate aminotransferase; CBC, complete blood count; EDSS, Expanded Disability Status Scale; EOT, end of treatment; IMP, investigational medicinal product; MSPS, Multiple Sclerosis Performance Scale; MusiQoL, Multiple Sclerosis International Quality of Life; PDDS, Patient-Determined Disease Steps; SAE, serious adverse event; SDMT, Symbol Digit Modalities Test; TSQM, Treatment Satisfaction Questionnaire for Medication.

Secondary Endpoints

- Change in TSQM score from baseline to Week 4 and to EOT in patients switching from another DMT.
- Change in TSQM from Week 4 to Week 48/EOT in treatment-naïve patients.
- Disease progression: change from baseline to Week 48/EOT on the PDDS scale.
- Disease progression: change from baseline to Week 24 and Week 48/EOT on the Multiple Sclerosis Performance Scale.
- Clinical outcomes: treated relapses, time to first treated relapse.
- Change in cognition as change from baseline to Week 48/EOT on the Symbol Digit Modalities Test.
- Occurrence of AEs.
- Adherence (ie, compliance) and persistence (ie, duration of exposure) to teriflunomide treatment over 48 weeks.
- QoL: change from baseline to Week 48/EOT measured by Multiple Sclerosis International Quality of Life and Stern Leisure Activity Scale.
- EDSS score at baseline and at Week 48/EOT.
- The timing of efficacy and safety assessments are shown in **Table 3**.

Analysis Populations

- Efficacy and safety populations will consist of all patients who receive at least one dose.

CONCLUSIONS

- Teri-PRO will improve clinical knowledge of the benefits of teriflunomide from a patient perspective with regard to efficacy, tolerability, and convenience.
- Teri-PRO will provide valuable information on PROs for MS disability progression, treatment satisfaction, adherence, QoL, and safety in patients with RMS receiving teriflunomide in routine clinical practice.

References

1. Gold R and Wolinsky J. *Acta Neurol Scand.* 2011;124:75-84.
2. Bruneau JM, et al. *Biochem J.* 1998;336(Pt 2):299-303.
3. Li L, et al. *Mult Scler J.* 2011;17:S422(P938).
4. O'Connor P, et al. *N Engl J Med.* 2011;365:1293-1303.
5. Kappos L, et al. *Mult Scler J.* 2012;18:9-53:54(153).
6. O'Connor P, et al. Presented at the International Society for Pharmacoeconomics and Outcomes Research, 2011.
7. Sanofi, data on file.
8. Vermesch P, et al. *J Neurol.* 2012;259:S1:1-236.
9. Atkinson MJ, et al. *Health Qual Life Outcomes.* 2004;2:12.
10. Hohol MJ, et al. *Neurology.* 1995;45:251-255.
11. http://www.narcoms.org/files/PDDS_Letter_111512_Final.pdf Accessed 14 May 2013.
12. Simeoni M, et al. *Mult Scler.* 2008;14:219-230.
13. Scarmeas N, et al. *Neurology.* 2001;57:2236-2242.

Disclosures

TV: Clinical Research (Biogen Idec, Genzyme, Jenssen Research, Teva Neuroscience, National Institutes of Health, Ono Pharmaceuticals), Consulting (Biogen Idec, Teva Neuroscience, Accelerated Cure Project, Genzyme, Bristol-Myers Squibb, Novartis Pharmaceuticals, Questco, Guthy-Jackson Charitable Trust, Xenoport, Acorda, Skolkovo Fund); ASB and MR: Employees of Genzyme, a Sanofi company.

Acknowledgments

Study supported by Genzyme, a Sanofi company.
Editorial assistance was provided by Marie Miller, Fishawack Communications, also funded by Genzyme, a Sanofi company.

Fifth Cooperative Meeting of CMSC and ACTRIMS, May 29–June 1, 2013, Orlando, FL, USA